A Phase II Open Label, Single-Blind, Multicenter, Single-Dose, Dose-Escalating Safety and Tolerability Study of Intrathecal Xen2174 in Oncology Patients

Author: Dr Lisa Stearns[1]; **Xenome Pain Study Group**: Prof. Michael Cousins[2], Dr Roger Gouke[3], Dr Tom Hughes[4], Dr Richard Rauck[5], Dr Miles Day[6], Dr Wendy Martin[7]

Affiliations: [1]Valley Cancer Pain Treatment Center, Scottsdale, AZ, USA; [2]Centre for Anaesthesia and Pain Management Research, Royal North Shore Hospital, St Leonards, NSW, Australia; [3]Department of Pain and Medicine, Sir Charles Gairdner Hospital, Nedlands, WA, Australia; [4]California Clinical Research, Inc., Davis, CA, USA; [5]Center for Clinical Research, Winston-Salem, NC, USA; [6]Anesthesiology/Pain Center, Texas Tech University Health Sciences Center, Lubbock, TX, USA, [7]Xenome Inc., San Diego, CA, USA

Introduction

Subject Objective: To evaluate the safety and tolerability of Xen2174, a synthetic conopeptide-based analgesic derived from venom peptide, MrIA. Study drug was administered as a single, intrathecal dose in a mixed oncology patient population who were either no longer responsive to, minimally responsive to, or intolerant of routine analgesic treatment.

Objectives of the protocol included: determination of maximum tolerated dose; safety and tolerability; pharmacokinetic profiling of plasma and CSF; and evaluation of pain response.

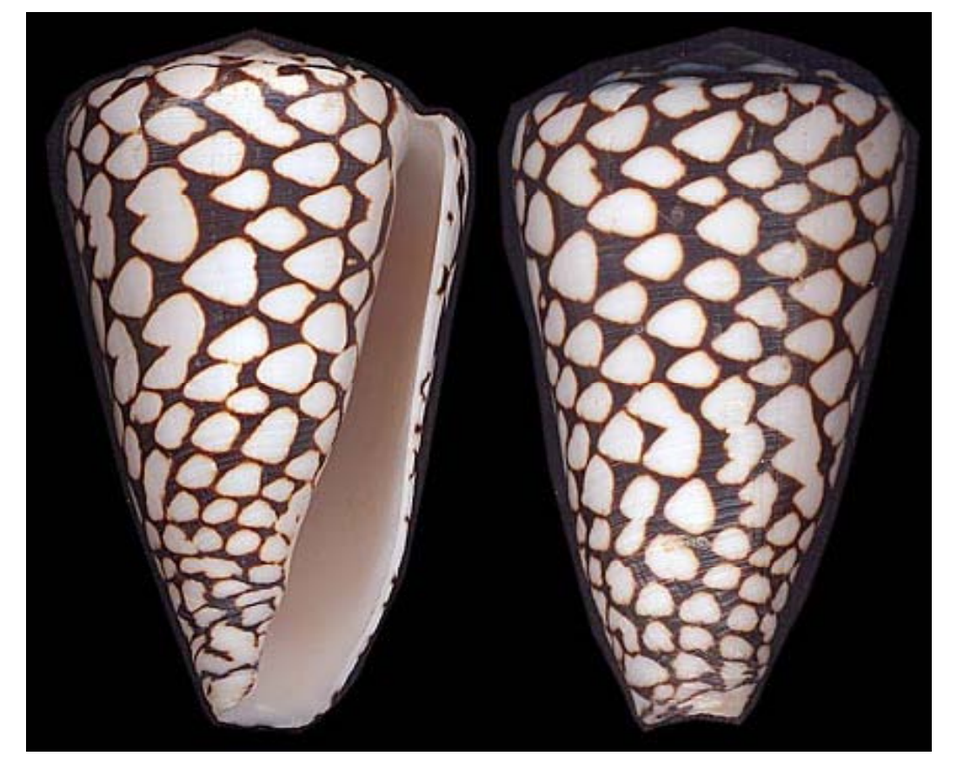

Figure 1. *Conus mamoreus*: natural source of MrIA (NGVCCGYKLCHOC)

Materials and methods

37 cancer patients with a history of chronic, intractable malignant pain were enrolled in this in-hospital study; the diagnosis of cancer could be ongoing or in remission. The primary endpoint was safety and tolerability of a single, bolus-dose administration of Xen2174, with CSF and plasma pharmacokinetics and efficacy analysis as secondary endpoints.

There was no restriction on type of prior oncology treatment, but only stable doses of oral chemotherapy could be co-administered with study drug. The chronic pain etiology was diverse, but similar in that it was not adequately treated with routine analgesia regimens.

Maximum Tolerated Dose (MTD) was assessed through a dose escalation design that included 10 completed dose levels, from 0.025mg through to 40mg (Table 1). Each dose escalation was monitored via a review of dose-limiting toxicities.

Safety was recorded through day 4 post dose via neurological and physical assessments, routine laboratory assessments, vital signs, and ECGs. Efficacy was recorded through day 4 post dose, primarily via patient rating of pain control and secondarily via global evaluation by study personnel.

Plasma pharmacokinetic sampling was obtained from dosing through day 2 post dose; 7 patients consented to optional CSF sampling.

Dose Strategy	Xen2174 (mg)	Patients Enrolled
Dose De-Escalation	0.025	1
	0.125	4
Starting Dose	0.25	4
Dose Escalation	0.75	6
	2.25	3
	5	3
	10	3
	20	3
	30	4
	40	6

Table 1. Summary of dose escalation strategy and number of patients by dose cohort

Results

Maximum Tolerated Dose

MTD was established at 30mg after 3 of 6 patients in the 40mg cohort experienced dose-limiting toxicities. These were:

- Apnea, Unresponsiveness, Grand Mal Seizure, Postictal State (1 pt.)
- Drug-Induced Aseptic Meningitis (1 pt.)
- Pain NOS (1 pt.)

Adverse Events

From the 37 patients enrolled in Study XC0205, there were a total of 41 treatment-emergent adverse experiences (AEs) across 17 patients (46%) with either a 'possible' (86%, 35 AEs from 14 patients), 'probable' (7%, 3 AEs from 2 patients) or 'definite' (7%, 3 AEs from 2 patients) relationship to the study drug, as assessed by the site investigators. Of these AEs, most were mild (CTCAE Grade 1: 56%, 23 AEs from 12 patients), moderate (CTCAE Grade 2: 22%, 9 AEs from 8 patients) or severe (CTCAE Grade 3: 20%, 8 AEs from 4 patients) in intensity (Table 2). One Grade 4 AE, dysphasia, was reported.

XC0205 Adverse Events

Relationship to Drug	CTC Grade					Total AEs
	1 (Mild)	2 (Moderate)	3 (Severe)	4 (Disabling)	5 (Death)	
1 Not Related	26	8	5		-	39
2 Unlikely	21	4	3	-	1	29
3 Possible	21	7	7	-	-	35
4 Probable	-	1	1	1	-	3
5 Definite	2	1	-	-	-	3
Total	70	21	16	1	1	109
All Related	23	9	8	1	0	41

Table 2. Adverse Events by CTCAE grading (severity) and relationship to study drug

Serious Adverse Events

In total there were 12 Serious Adverse Events (SAEs) across 5 patients from various dose cohorts. 7 SAEs from 3 patients were considered possibly, probably or definitely related to the study drug. Of these 7 related events, 4 events from 2 patients were classified as nervous system disorders (Unresponsiveness, Grand Mal Seizure, Postictal State – 1 patient; Dysphasia – 1 patient). The remaining SAEs were split across Infections (Drug-Induced Aseptic Meningitis – 1 patient), Psychiatric disorders (Confusion – 1 patient), Respiratory disorders (Apnea – 1 patient) (Figure 2). Two of the 3 patients with related SAEs were from the 40mg dose cohort.

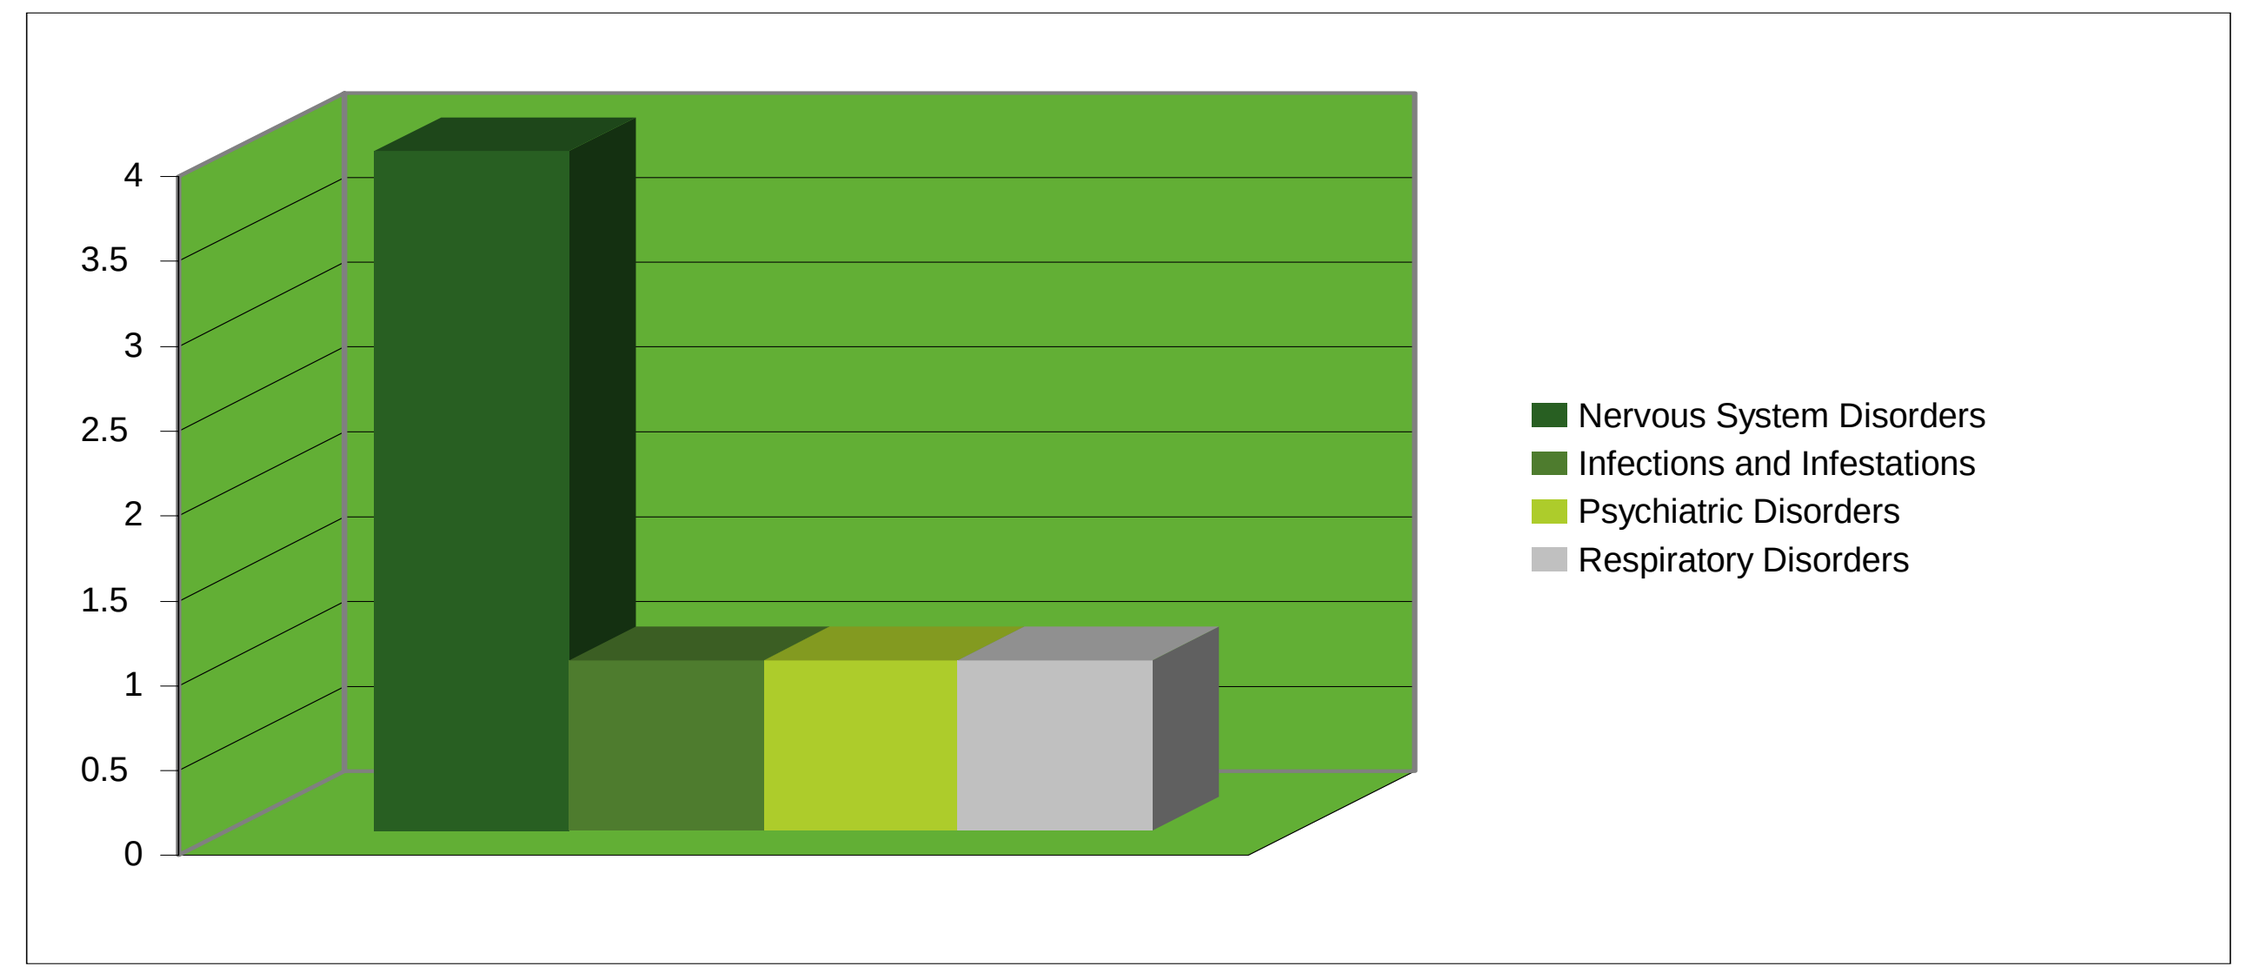
4
3.5
3
2.5
2
1.5
1
0.5
0
Nervous System Disorders
Infections and Infestations
Psychiatric Disorders
Respiratory Disorders

Figure 2. Drug-related serious adverse events by body system classification

Pharmacokinetics

Plasma PK analysis showed that C_{max} and AUC_{inf} values increased with increase in dose, in an approximately dose-proportional manner (Figure 3). Maximum plasma concentrations occurred at 4 to 10 hours after dosing and the apparent terminal-phase half lives ranged from about 10 to 24 hours. Overall, the pharmacokinetic profile of Xen2174 in plasma likely represents the rate of transfer from the CSF into the plasma, rather than the elimination of the drug by systemic clearance processes.

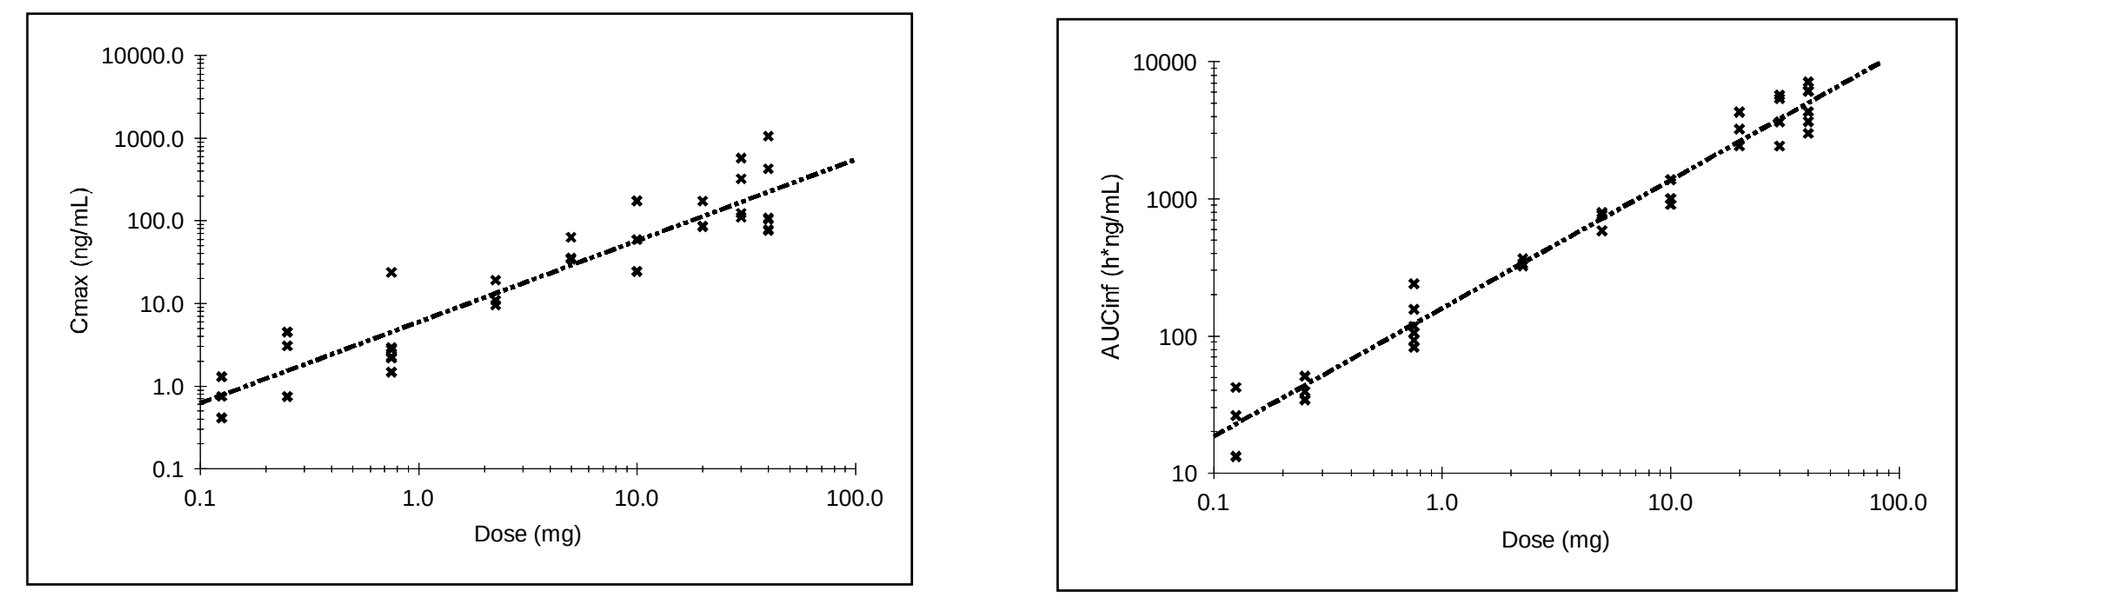
Cmax (ng/mL)
10000.0
1000.0
100.0
10.0
1.0
0.1
Dose (mg)
AUCinf (h*ng/mL)
10000
1000
100
10
0.1
1.0
10.0
100.0

Figure 3. Relationship between Xen2174 (a) C_{max} and (b) AUC_{inf} and Dose (Plasma)

CSF PK analysis on a subset of 7 patients showed that exposure increased with increase in dose. Maximum CSF concentrations occurred at 1 hr post dose with the exception of one patient with a T_{max} of 8 hours. The terminal-phase half lives ranged from about 2 to 14 hours and likely reflect the rate of transfer of Xen2174 from the CSF to plasma.

Efficacy

Based on analysis of the efficacy assessments (and excluding the NOEL cohort of 0.025mg), inferential statistics revealed the following:

Changes in Individual patient pain scores from pre-dose baseline:
- Every cohort experienced at least 1 patient with >90% improved primary pain relief during the assessment period
- Cohorts 0.125mg, 0.75mg, 2.25mg, 5mg, 10mg, 30mg, 40mg all contained at least 1 patient who had 100% primary pain relief during the assessment period

Group mean percent change in patient pain scores from pre-dose baseline :
- Peak pain relief for all dose cohorts was observed between 36 - 96hrs post dose
- All cohorts experienced a reduction in their primary pain at the following time points: 1hr, 2hr, 4hr, 8hr, 12 hr, 24hr, 48hr, and 96hr
- All cohorts experienced >45% improvement in their primary pain scores across at least 1 time point within the first 12 hours post dose

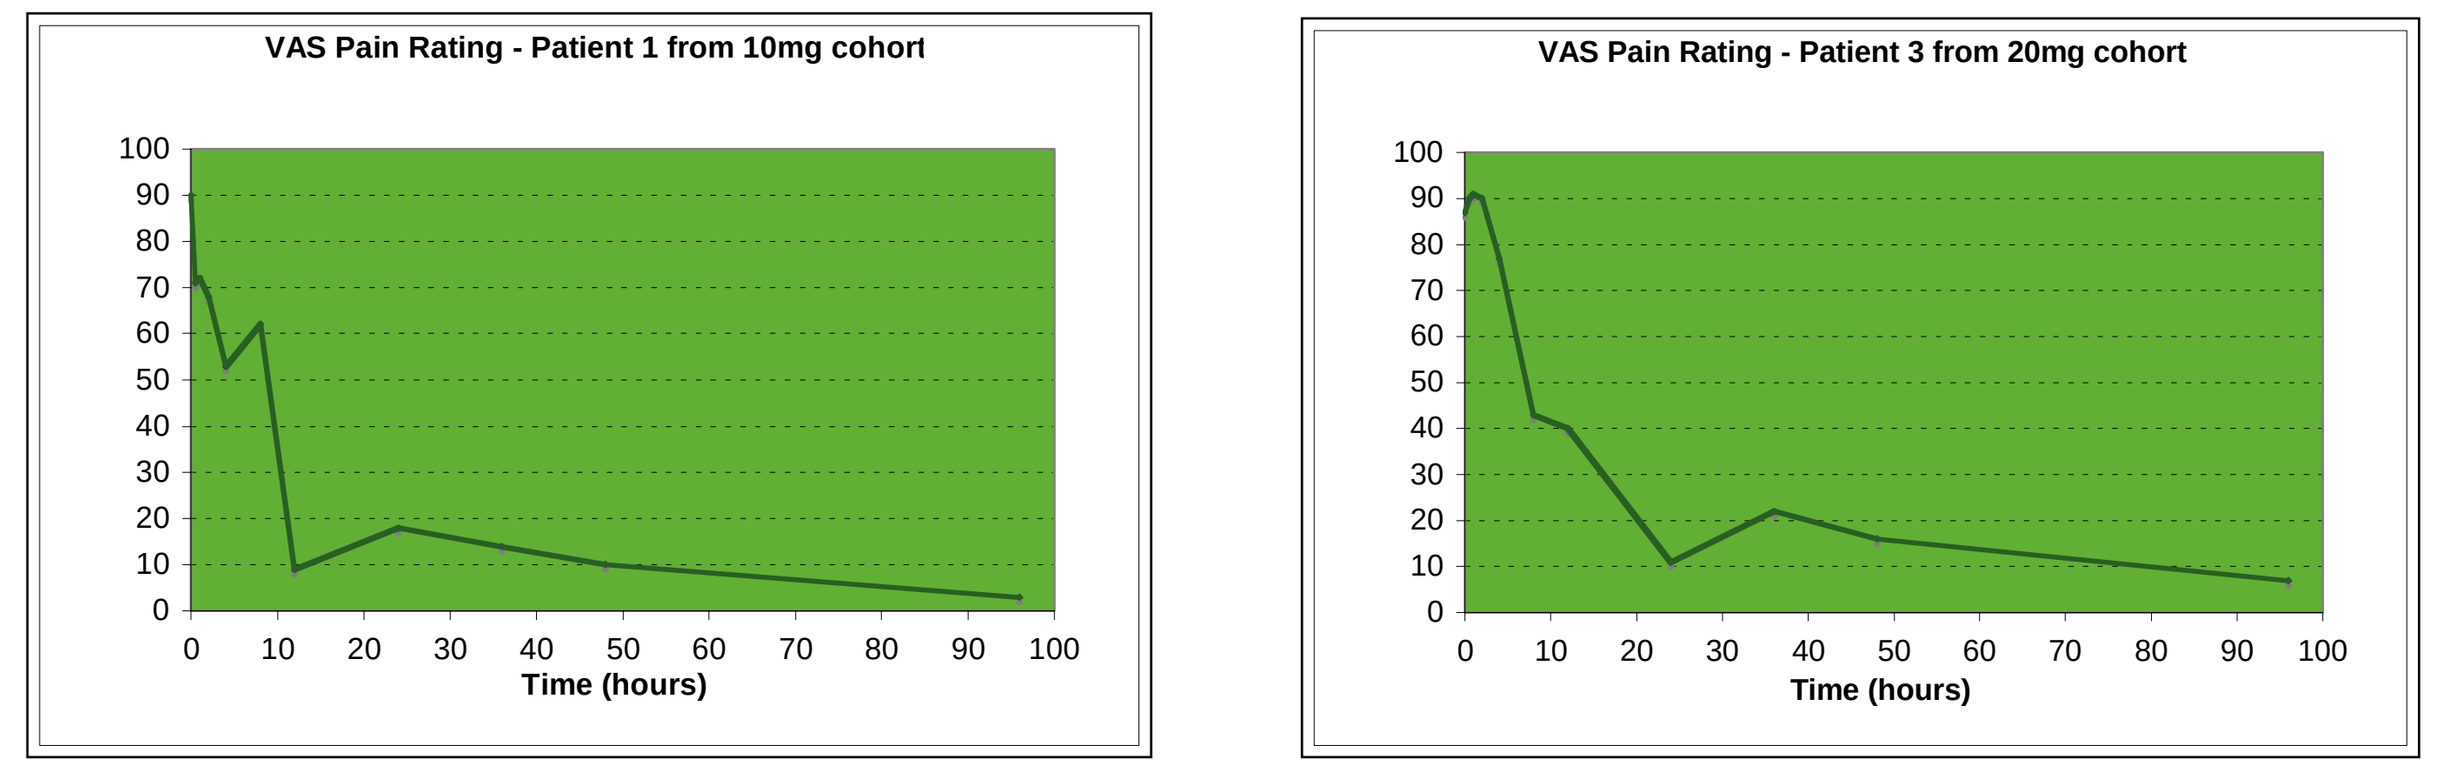
VAS Pain Rating - Patient 1 from 10mg cohort
VAS Pain Rating - Patient 3 from 20mg cohort
100
90
80
70
60
50
40
30
20
10
0
0 10 20 30 40 50 60 70 80 90 100
Time (hours)

Figure 4. Two examples of patient VAS assessment results (100 point scale), based on the question: How would you assess your current level of cancer pain?

Conclusions

- A single bolus intrathecal injection in a mixed oncology patient population showed Xen2174 to be both safe and tolerated in a dose range from 0.025mg through to and including 30mg
- Adverse event analysis identified no consistent trends in side effects
- 78% of all drug related, treatment emergent adverse events were considered either mild or moderate in severity
- Of the 12 SAEs experienced, 7 SAEs (17% of all related AEs) from 3 patients were considered related to the study drug. Two of the 3 patients were from the 40mg cohort
- PK analysis revealed that both C_{max} and AUC_{inf} values increased in a dose-proportional manner
- Efficacy results are indicative of both early onset and extended duration of action, making Xen2174 an ideal candidate for continued clinical development

Background Publications

Nielsen CK, Lewis RJ, Alewood D, et al. Anti-allodynic efficacy of the chi-conopeptide, Xen2174, in rats with Neuropathic pain. Pain. 2005 Nov; 118(1-2): 112-24

Obata H, Conklin D, Eisenach JC. Spinal noradrenaline transporter inhibition by reboxetine and Xen2174 reduces tactile hypersensitivity after surgery in rats. Pain. 2005 Feb; 113(3):271-6

Sharpe IA, Palant E, Schroeder CI, et al. Inhibition of the norepinephrine transporter by the venom peptide chi-MrIA. Site of action, Na+ dependence, and structure-activity relationship. J Biol Chem 2003 Oct; 278(41):40317-23

Sharpe IA, Gehrmann J, Loughnan ML, et al. Two new classes of conopeptides inhibit the alpha 1-adrenoceptor and noradrenaline transporter. Nat Neurosci 2001 Sep; 4(9):902-7

For further information

Please contact Xenome via email at enquiries@xenome.com or by phone on +61 7 3720 8055.

More information on this and related projects can be obtained via the company's website, www.xenome.com .